Institutional Repository - Research Portal
Dépôt Institutionnel - Portail de la Recherche
researchportal.unamur.be

University of Namur

RESEARCH OUTPUTS / RÉSULTATS DE RECHERCHE

Inter-laboratory variability of the standardized ETP-based APC resistance assay (PB0386)

Morimont, Laure; Maloteau, Vincent; Devel, Philippe; Dogné, Jean-Michel; Bouvy, Céline; Douxfils, Jonathan

Publication date:
2020

Link to publication

Citation for pulished version (HARVARD):
Morimont, L, Maloteau, V, Devel, P, Dogné, J-M, Bouvy, C & Douxfils, J 2020, 'Inter-laboratory variability of the standardized ETP-based APC resistance assay (PB0386)', ISTH 2020 virtual congress: July 12-14, 2020, 12/07/20 - 14/07/20.

General rights
Copyright and moral rights for the publications made accessible in the public portal are retained by the authors and/or other copyright owners and it is a condition of accessing publications that users recognise and abide by the legal requirements associated with these rights.

• Users may download and print one copy of any publication from the public portal for the purpose of private study or research.
• You may not further distribute the material or use it for any profit-making activity or commercial gain
• You may freely distribute the URL identifying the publication in the public portal ?

Take down policy
If you believe that this document breaches copyright please contact us providing details, and we will remove access to the work immediately and investigate your claim.

Download date: 27. Jul. 2024

Inter-laboratory variability of the standardized ETP-based APC resistance assay (PB0386)

L. MORIMONT [1,2], V. Maloteau [2], P. Devel[2], J-M. Dogné[2], C. Bouvy[1], J. Douxfils[1,2]

[1] QUALIblood s.a., Namur Belgium

[2] University of Namur, Department of Pharmacy, Namur Thrombosis and Hemostasis center (NTHC), Namur Research Institute for Life Sciences (NARILIS), Namur, Belgium

ISTH 2020 JULY 12-14 VIRTUAL CONGRESS ISTH2020.ORG The science of today is the innovation of tomorrow

QUALIblood Your expert in blood testing

UNIVERSITÉ DE NAMUR

FÉDÉRATION WALLONIE-BRUXELLES

INTRODUCTION

- Risk minimization measures need to be implemented to reduce the thrombotic risk associated with the use of combined oral contraceptives (COC). However, there is controversy on how to assess the risk at an acceptable cost for the society.
- Despite regulatory bodies recommend the assessment of the ETP-based activated protein C (APC) resistance during the development of steroid contraceptives; results are not comparable between studies due to lack of validation and standardization.
- To overcome this issue, our group recently validated and standardized this assay.
- However, further investigations are needed before a wide implementation of this method.

AIM

To assess the inter-laboratory variability of the validated ETP-based APC resistance assay.

METHOD

- Analyses were performed on 3 Calibrated Automated Thrombogram (CAT) with Thrombinoscope software (version 5.0 for $CAT_{1\&2}$; version 2.0 for CAT_3) and by 3 different operators.
- Dose-response curves (n=3) were performed on each device to defined the amount of APC, leading to
 - (a) 90% of inhibition of the ETP on an in-house reference plasma,
 - (b) the corresponding inhibition % on a commercial reference plasma.
- Quality controls (n=3) and a subset of real-life samples (n=11) were tested on each equipment.
- All measurements were performed in duplicate.

RESULTS

- Dose-response curves did not show any significant differences between CAT_1 and CAT_2 (Dunn's multiple comparison test; p-value > 0.9999) on both, commercial and in-house reference plasma ▶**Table/Figure 1.**
- An APC concentration of 217 mU/mL led to 90% of inhibition of the ETP on the homemade reference plasma and 68% of inhibition on the commercial reference plasma ▶**Table/Figure 1**.

FIGURE 1: Total variability between CAT_1 and CAT_2. The red dotted line represents the reference range of in-house reference plasma (90±2.5%) and the blue dotted line represents the reference range of commercial reference plasma (68±8%). The green dotted line represents the defined concentration of APC (217 mU/mL) to fit the reference ranges.

TABLE 1: Inhibition percentage at 6 concentration levels of APC with in-house reference plasma and commercial reference plasma on CAT_1 and CAT_2. Inhibition % in blue are outliers*

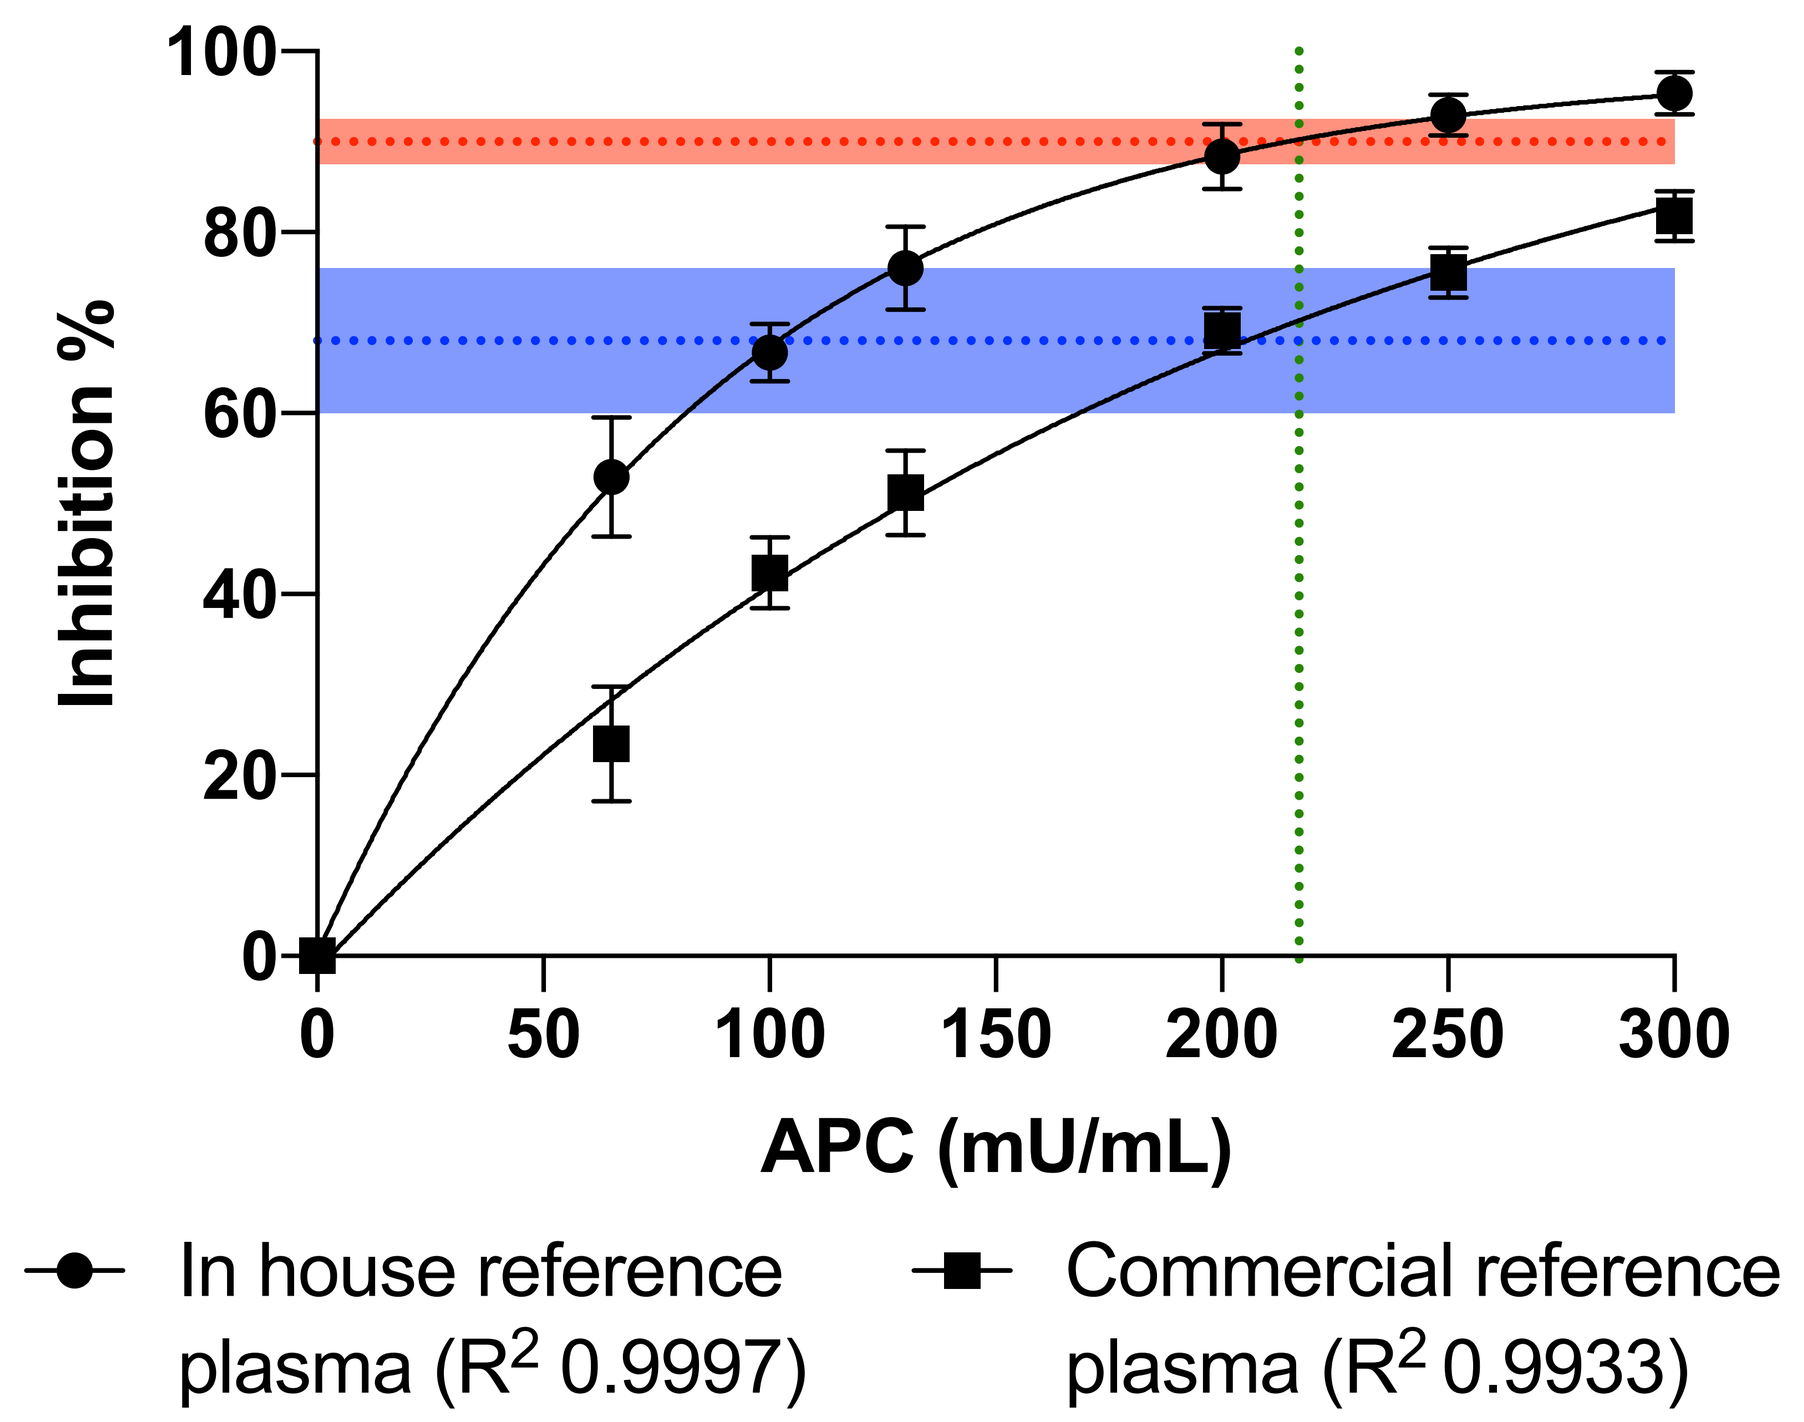
Inhibition %
100
80
60
40
20
0
0
50
100
150
200
250
300
APC (mU/mL)
In house reference plasma (R^2 0.9997)
Commercial reference plasma (R^2 0.9933)

[APC]	In-house reference plasma: Inhibition % CAT 1	CAT 2	p-value	Commercial reference plasma: Inhibition % CAT1	CAT2	p-value
65 mU/mL	26,69*	52,05	>0.9999	14,65	27,48	>0.9999
	28,71*	56,13		17,18	31,35	
	44,24	59,57		25,53	24,7	
100 mU/mL	64,77	70,39	>0.9999	38,94	40,71	>0.9999
	62,63	64,34		37,51	47,3	
	69,72	68,21		44,32	45,7	
130 mU/mL	73,9	80,1	>0.9999	46,84	52,14	>0.9999
	68,15	76,11		44,37	56,42	
	80,23	77,84		52,77	54,82	
200 mU/mL	90,27	89,45	>0.9999	65,05	67,54	>0.9999
	87,92	81,62		68,86	71,88	
	92,09	88,72		71,27	69,99	
250 mU/mL	95,25	93,42	>0.9999	74,02	74,4	>0.9999
	91,58	89,48		72,16	78,83	
	95,47	92,47		79,06	74,66	
300 mU/mL	97,19	95,87	>0.9999	78,86	79,58	>0.9999
	96,13	82,89*		83,06	85,8	
	96,55	95,08		83,69	79,92	
Defined concentration of APC (95%CI): **217 mU/mL** [197 mU/mL-249 mU/mL]						

- A mean deviation of 7.83% was observed when comparing samples tested on both CAT device at the defined APC concentration of 217 mU/mL (▶**Table 2**).

TABLE 2: Inhibition percentage and absolute difference between CAT1 and CAT2.

	Inhibition % CAT 1	Inhibition % CAT 2	Difference
In-house ref plasma	88,66%	94,56%	5,90%
Commercial ref plasma	64,81%	74,36%	9,55%
QC Low	98,19%	100,16%	1,96%
QC Norm	31,22%	44,17%	12,95%
QC High	7,18%	9,70%	2,52%
S1	91,99%	96,34%	4,36%
S2	77,01%	81,54%	4,53%
S3	78,53%	86,07%	7,54%
S4	64,74%	74,16%	9,42%
S5	56,94%	64,92%	7,97%
S6	68,84%	79,33%	10,49%
S7	69,03%	76,53%	7,49%
S8	69,02%	76,42%	7,40%
S9	46,88%	60,55%	13,67%
S10	61,99%	71,36%	9,37%
S11	40,69%	50,90%	10,21%
	Mean deviation		7,83%

- As the software version of the CAT_3 was older, some modifications must be performed to ensure the transferability of the methodology. This is due to important background noise which limits the performance of this older version of the CAT_3 at low ETP-values.

CONCLUSIONS

- These results demonstrate the transferability of the validated ETP-based APC resistance test between CATs using the same software version.
- In order to confirm the reliability of the results obtained with this validated method, its implementation in other centre is essential.
- The conduct of a larger multi-centre study should therefore be performed before implementing this technique in the routine setting.

REFERENCES

[1] Guideline on clinical investigation of steroid contraceptives in women - EMEA/CPMP/EWP/519/98 Rev 1.

[2] Morimont, L., et al. (2020). "Proof of concept of a new scale for the harmonization and the standardization of the ETP-based APC resistance." J Thromb Haemost **18**(4): 895-904.

[3] Douxfils, J., et al. (2020). "Validation and standardization of the ETP-based activated protein C resistance test for the clinical investigation of steroid contraceptives in women: an unmet clinical and regulatory need." Clin Chem Lab Med **58**(2): 294-305.

ACKNOWLEDGEMENTS

This study was financed by QUALIblood s.a and the brussels-Wallonia federation (convention no. 8031).

CONTACT INFORMATION

Email: laure.morimont@qualiblood.eu

Tel: 32 (0)81 72 42 92